Poster # 006

SUBANALYSIS OF AN OPEN-LABEL, CROSSOVER STUDY TO ASSESS THE RELATIVE BIOAVAILABILITY OF SELF-ADMINISTERED NASAL EPINEPHRINE COMPARED TO ADMINISTRATION BY TRAINED HEALTH PERSONNEL IN HEALTHY ADULT SUBJECTS

David Dworaczyk, PhD,[1] and Allen Hunt, MD[2]
[1]Bryn Pharma, LLC, Raleigh, NC, USA; [2]Celerion, Lincoln, NE, USA

INTRODUCTION

- Anaphylaxis is a situation where a person experiences a serious acute allergic reaction that requires immediate attention to avoid serious morbidity and mortality
- Epinephrine is the first-line therapy for anaphylaxis, commonly administered via intramuscular (IM) autoinjector injection[1]
- Self-administration of injections has an associated social stigma that can lead to treatment hesitancy[2,3]
- Patient adherence with autoinjector use may be compromised owing to patient lack of compliance to carry their autoinjectors with them routinely, reluctance to use self-injectors (eg, needle anxiety or fear) or application error (eg, lack of training, injection injuries)[1–7]
- Delayed epinephrine administration or exposure during anaphylactic events may increase risk of hospitalizations and potentially fatal outcomes[8]
- Although this is a nasal spray and not an inhaler, inhaled therapeutics are well accepted for several acute interventions requiring rapid onset of action and self-administration, such as those for bronchodilation and reversal of opioid overdose[9-12]
- Intranasal (IN) administration is under development for the treatment of anaphylaxis[9–12]
- The IN route, therefore, represents an attractive potential alternative to an autoinjector for the treatment of patients experiencing an anaphylactic event[9]

RATIONALE

NDS1C is a self-administered, IN dosage form of epinephrine intended for the treatment of allergic reactions (type1) and is currently being evaluated as a potential novel therapeutic option in the treatment of patients experiencing anaphylactic events. Data previously presented[13] suggest that the Bryn nasal spray may produce a more favorable epinephrine exposure profile and alleviation of anaphylactic symptoms as compared with the epinephrine autoinjector. However, in trials conducted to date, doses have been administered by trained professionals (TPs). The present study compares the pharmacokinetic (PK) profile of NDS1C 6.6 mg when TP-administered to NDS1C 6.6 mg when self-administered, hypothesizing that the ease of use would enable appropriate administration, providing comparable PK profiles in both scenarios.

OBJECTIVES

- The objective of this study was as follows:
- To assess the outcome of a single dose of TP-administered IN epinephrine to that of self-administered IN epinephrine in healthy subjects based on the following parameters:
 - Primary endpoint
 - Comparative PK and relative bioavailability (BA)
 - Secondary endpoints
 - Comparative pharmacodynamics (PD) expressed as changes in blood pressure (BP) and heart rate (HR)
 - To assess the safety and tolerability of epinephrine following administration by nasal spray in healthy subjects

METHODS

A phase 1, open-label, crossover design study was conducted to assess the relative bioavailability of TP- or self-administered IN epinephrine in healthy adult volunteers. A total of 83 adult male and female subjects were enrolled and 80 subjects completed the study. Screening of subjects occurred within 28 days prior to the first dosing. Subjects were randomly assigned to two groups: group A, TP-administered and group B, self-administered. Each subject acted as their own control. All 83 subjects enrolled were included in the PK and safety analyses.

STUDY PARTICIPANTS

Treatment groups were as follows:
- Treatment A:
 - 6.6 mg epinephrine (0.11 mL x 60 mg/mL NDS1C) administered IN by a TP at Hour 0 on Day 1
- Treatment B:
 - 6.6 mg epinephrine (0.11 mL x 60 mg/mL NDS1C) self-administered IN at Hour 0 on Day 1

DOSE ADMINISTRATION

Subjects blew their nose immediately prior to initiation of dosing in each IN treatment to clear their nostrils. The device was held with thumb at the base of the device and spray nozzle between two fingers. The TP (Treatment A) or subject (Treatment B) closed one nostril with his/her finger and inserted the tip of the bottle into the other nostril, aiming towards the back of the nose as straight as possible (spray nozzle was not aimed toward nasal septum). The drug was administered while subjects were breathing gently through their nose, and gently sniffed after a dose was administered to keep any of the dose from running out of the nose. Subjects were asked to try and refrain from sneezing or nose blowing for 30 minutes following nasal administration. Events of sneezing and/or blowing of the nose within 30 minutes of administration were recorded.

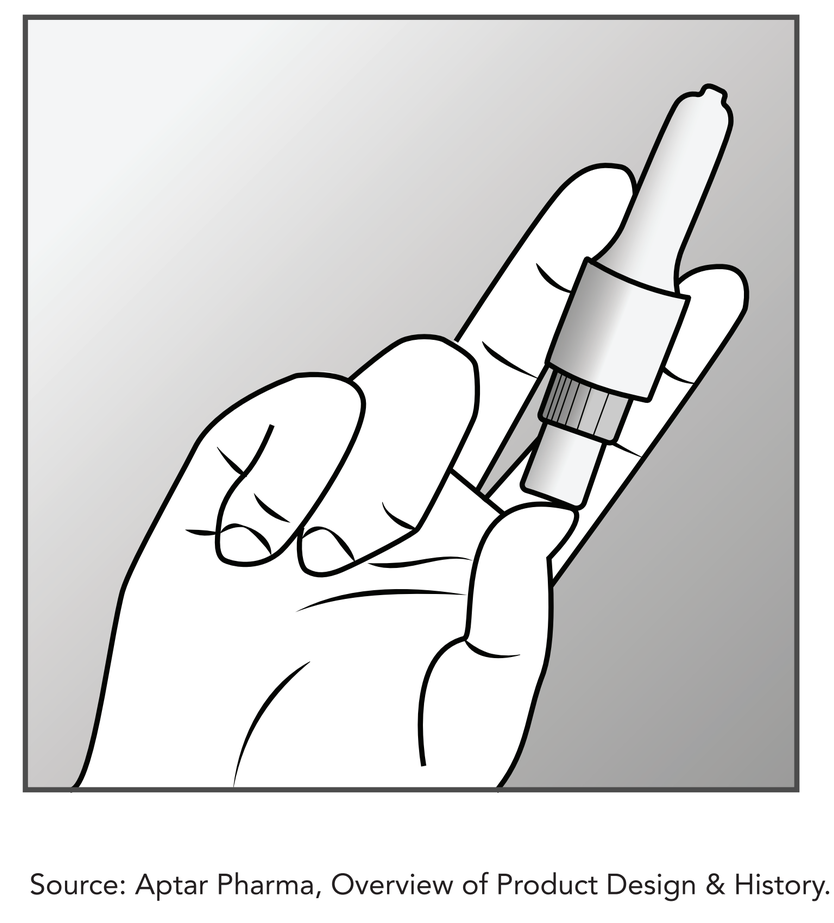
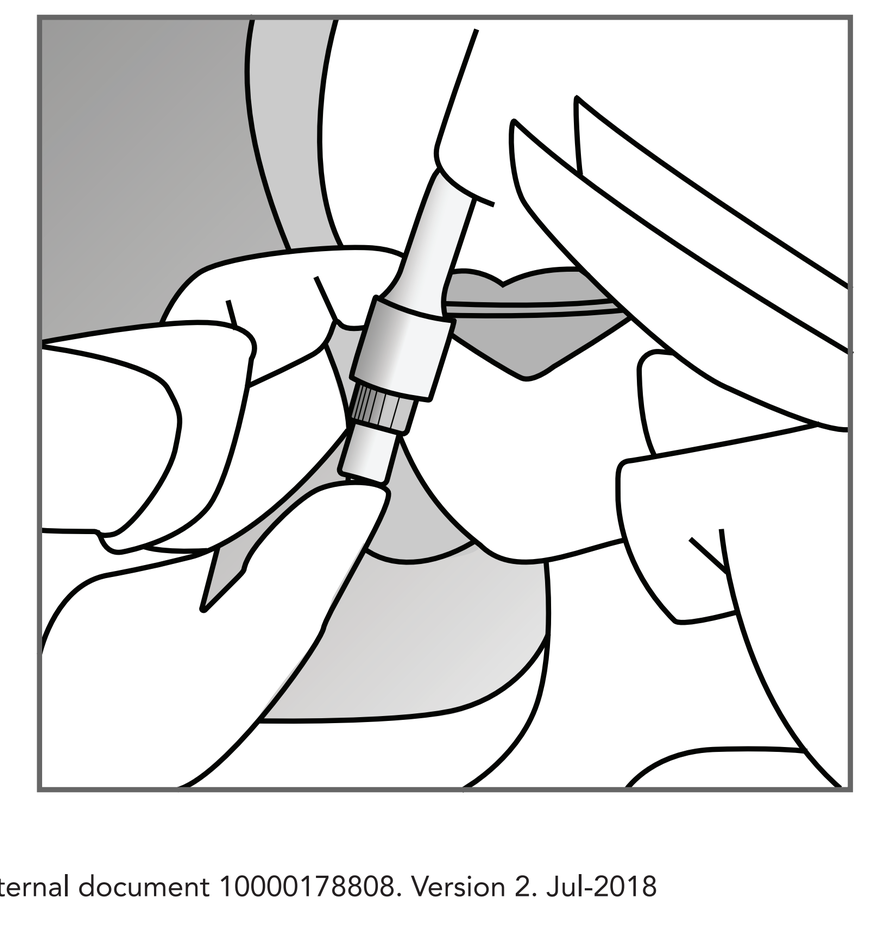

Source: Aptar Pharma, Overview of Product Design & History. Internal document 10000178808. Version 2. Jul-2018

STUDY ASSESSMENTS

The primary endpoint of the study was bioavailability, with secondary endpoints evaluating pharmacodynamics (HR and BP) and relative safety. Baseline assessments were conducted to correct for endogenous epinephrine levels and blood samples were taken over a six-hour period for assessment of plasma epinephrine. Emphasis was placed on the PK results within the first 60 minutes.

Pharmacokinetics
Serial blood samples for the determination of plasma epinephrine were collected at -30, -20, and -10 minutes pre-dose and 1, 3, 5, 7, 10, 15, 20, 25, 30, 45, 60, 90, 120, 180, and 360 minutes post-dose.

Pharmacodynamics
HR and BP were measured at -30, -20, and -10 minutes pre-dose and 1, 3, 5, 7, 10, 15, 20, 25, 30, 45, 60, 90, 120, 180, and 360 minutes post-dose.

Safety
Safety was evaluated by clinical laboratory tests, physical examination, vital signs, 12-lead electrocardiograms (ECGs), and adverse events (AEs).

STATISTICAL ANALYSES

Since this was a crossover design, the statistical comparison of Group A versus Group B was completed using a subset of the available PK parameter data. The comparison utilized the PK parameters from only the first IN administration to control for any potential carryover effects from the first to the second doses. The aim was to determine whether the first IN administration could be considered equivalent, with the geometric mean ratio falling within the range of 80.00 – 125.00%. This ANOVA was performed using SAS® PROC MIXED.

RESULTS

PHARMACOKINETICS

- Comparable PK/PD profiles were observed when IN epinephrine was TP- or self-administered. The geometric LSMs for AUC $T_{0\text{-}360}$ (min*pg/mL) were 17,748 (A) and 18,273 (B) with intra-subject %CV=52.19. The observed C_{max} (pg/mL) was 197.32 (A) and 182.98 (B), with intra-subject %CV=75.61. All other time points demonstrated similar consistency among the cohorts. Geometric mean T_{max} was achieved by 21.63 (A) and 19.82 min (B). Comparable number/percent of subjects achieved ≥100 pg/mL within 60-min post dose, regardless of TP- or self-administration.
- In general, the baseline-adjusted PK parameters were similar for the 6.6-mg IN formulation whether TP- or self-administered, demonstrating that the BA for IN-administered epinephrine was equivalent regardless of the administrator.

PHARMACODYNAMIC RESULTS

- TP-administered IN epinephrine and self-administered IN epinephrine demonstrated mean unadjusted and baseline-adjusted vital sign values for HR, SBP, and DBP within normal limits with no remarkable differences observed in these values.
- Mean HR baseline-adjusted values did not exceed ±10 bpm at any post-dose time point, mean SBP baseline-adjusted values for TP- and self-administered IN epinephrine were 1.2 + 19.2 mmHg and 1.5 + 15.77 mmHg, respectively, and mean DBP baseline-adjusted values were 7.0 + 10.08 mmHg and 4.7 + 9.50 mmHg in both administration groups.
- Overall, there was no trend observed in statistical comparisons for AUEC0-t and E_{max} for baseline-adjusted vital sign parameters. However, the difference in baseline-adjusted DBP values for TE_{max} between 6.6 mg epinephrine IN self-administered versus 6.6 mg epinephrine IN TP-administered was found to be statistically significant ($P = 0.0465$).

SAFETY RESULTS

- There were minimal differences in reported AEs among treatment groups. Mild administration site pain (22%) was the most frequently reported event following administration of 6.6 mg epinephrine IN (TP-administered) and following 6.6 mg epinephrine IN (self-administered). Most events were considered by the PI to be mild, and all events resolved during the study.
- Overall, there were no remarkable safety findings in the safety assessments for vital signs, ECGs, and clinical laboratory parameters.

Baseline-Adjusted Plasma Epinephrine Concentrations (pg/mL) Following first IN Administration of 6.6 mg Epinephrine (0.11 mL x 60 mg/mL NDS1C) Intranasal spray (pharmacokinetic population)

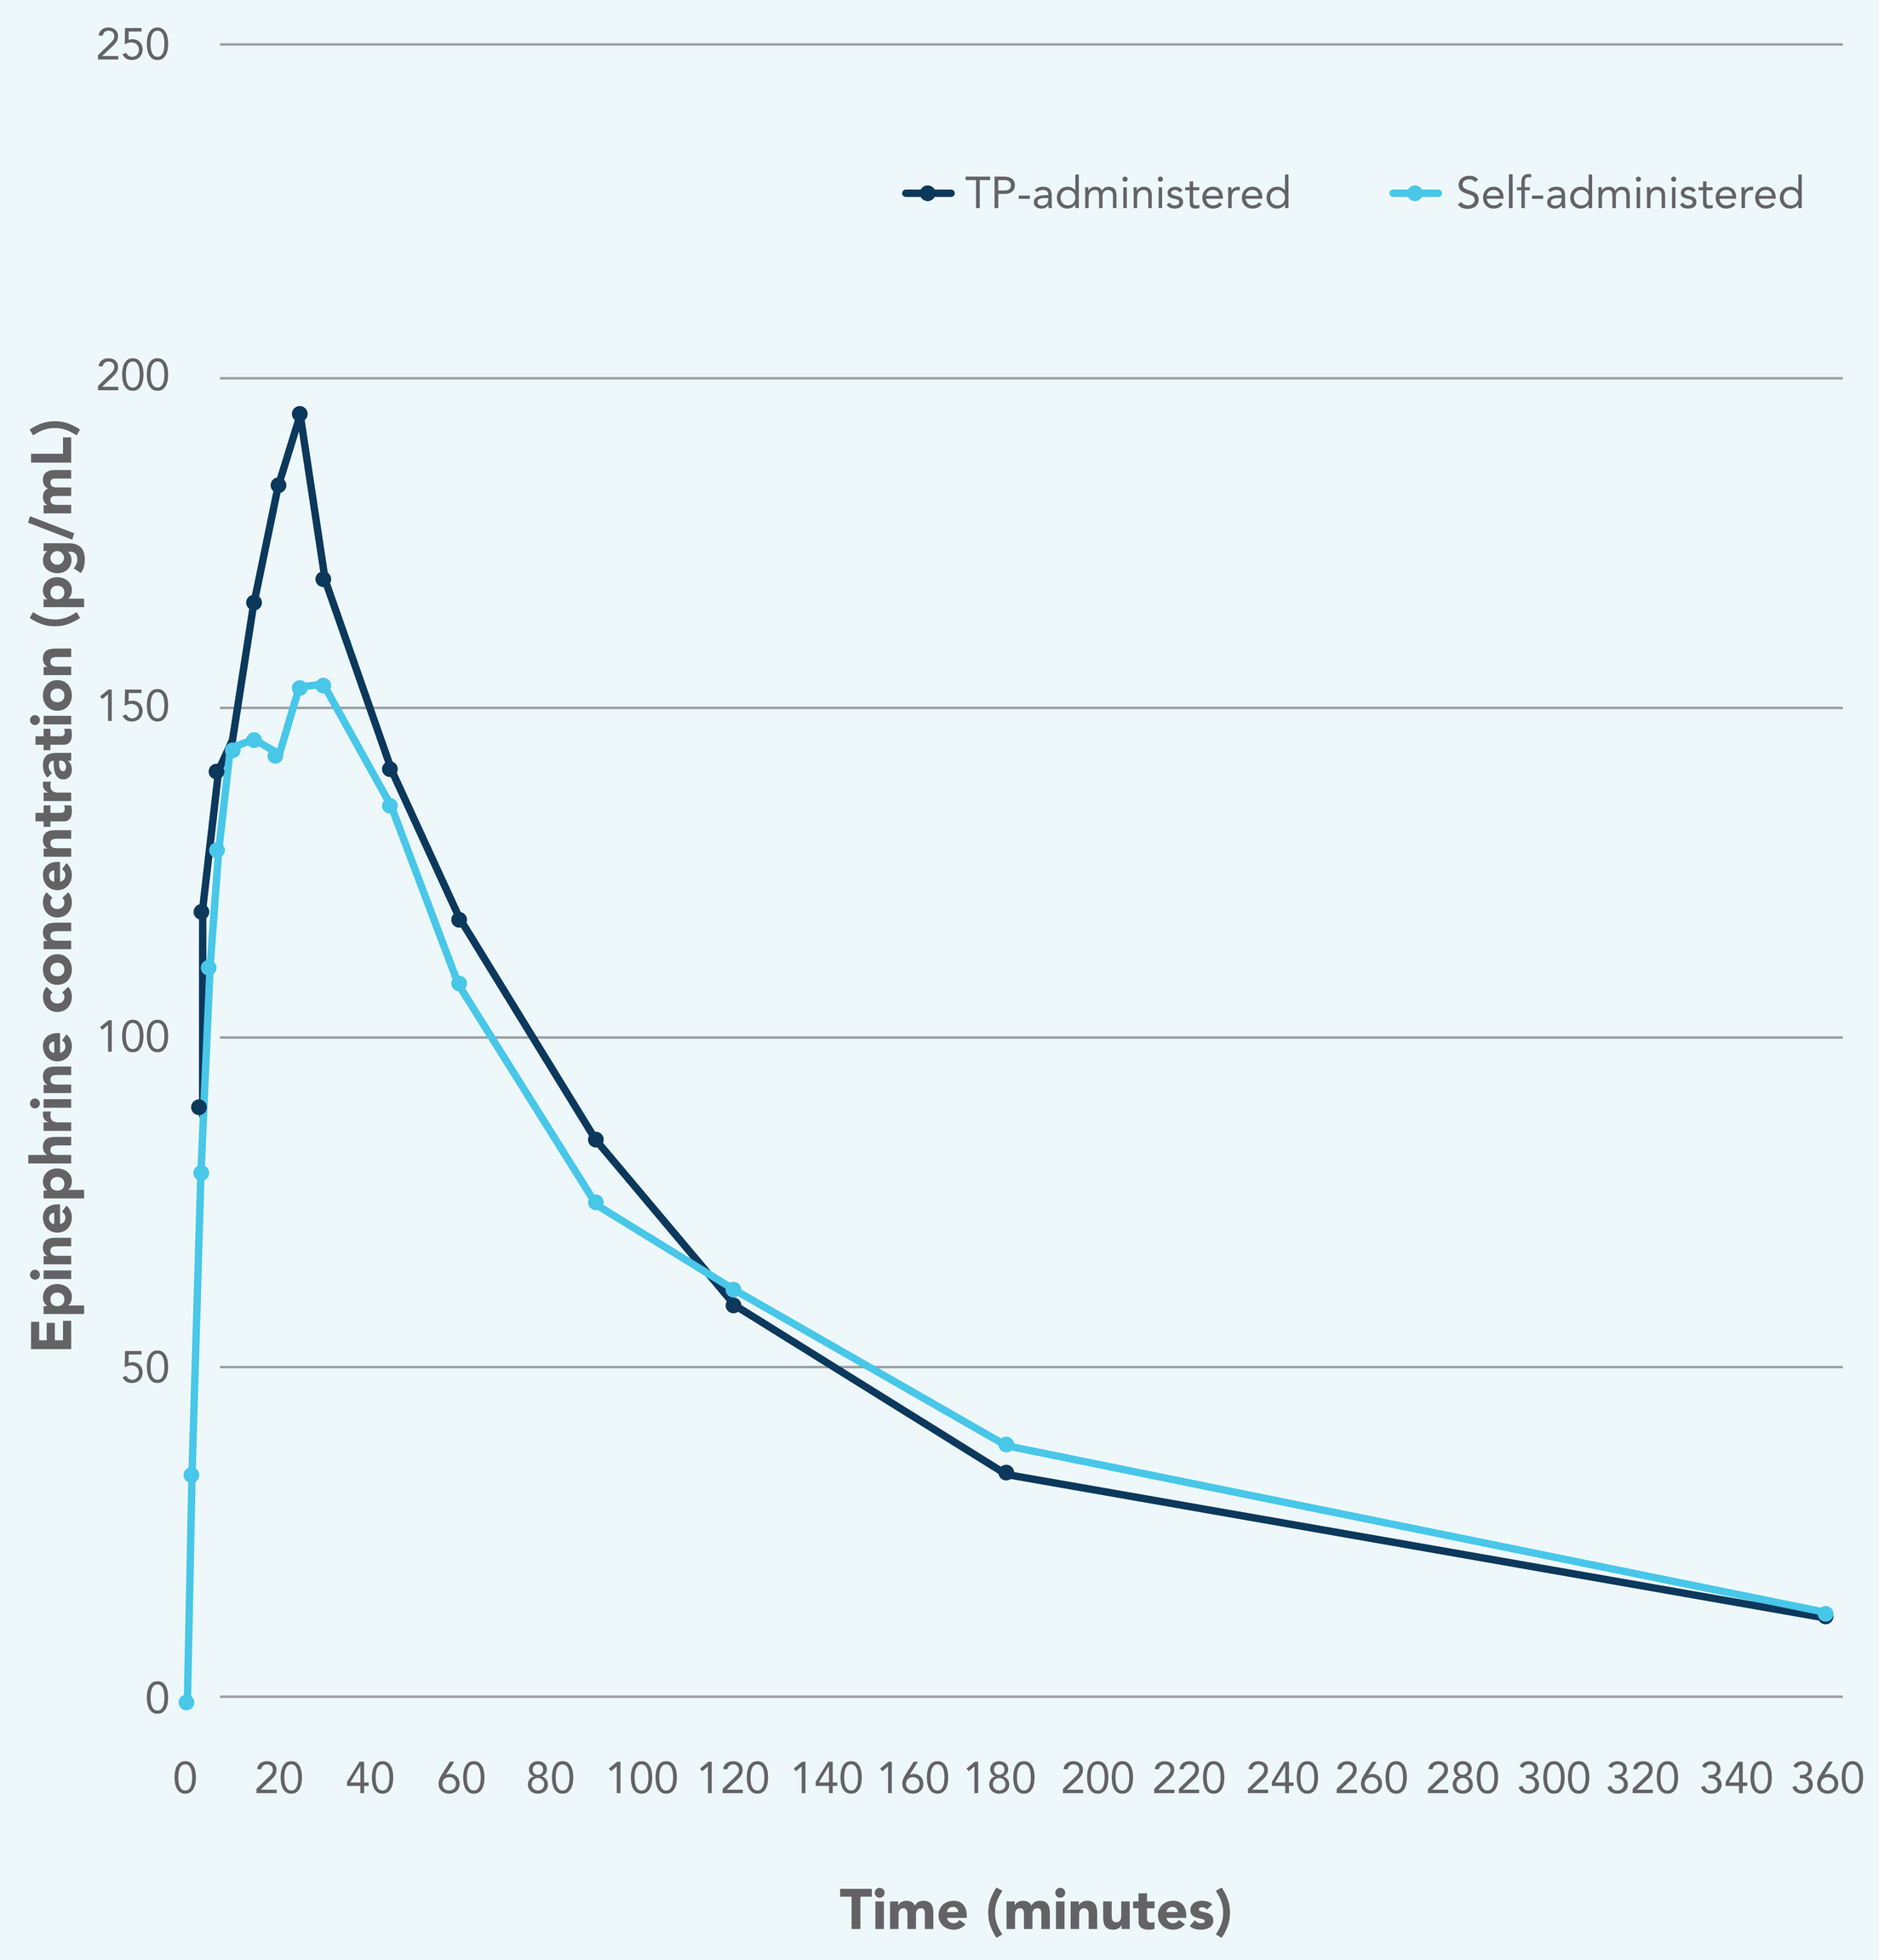
TP-administered
Self-administered
Epinephrine concentration (pg/mL)
250
200
150
100
50
0
0 20 40 60 80 100 120 140 160 180 200 220 240 260 280 300 320 340 360
Time (minutes)

ACKNOWLEDGMENTS AND DISCLOSURES: This research was supported by Bryn Pharma, LLC. Medical writing assistance was provided by Alden Masonis, PhD, and Donna Schroeder, PharmD, of ODIN's Strategic Solutions; funding for this assistance was provided by Bryn Pharma, LLC. David Dworaczyk, PhD, is an employee of Bryn Pharma, LLC. Allen Hunt, MD, is an employee of Celerion and received funding support from Bryn Pharma, LLC, to conduct this research study.

REFERENCES: **1.** Posner LS, Camargo CA Jr. *Drug Healthc Patient Saf.* 2017;9:9–18. **2.** Niño de Guzmán Quispe E, Martínez García L, Orrego Villagrán C, et al. *Patient.* 2021;14(6):719-740. **3.** Uhm JY, Choi MY. *Healthcare (Basel).* 2020;8(4):506. **4.** Bellanti JA, Settipane RA. *Allergy Asthma Proc.* 2014;35(3):195–196. **5.** Simons FER, Ebisawa M, Sanchez-Borgas M, et al. *World Allergy Organ* J. 2015;8(1):32. **6.** McLenon J and Rogers MAM. *J Adv Nurs.* 2019;75(1):30–42. **7.** Portnoy J, Wade RL, Kessler C. *J Allergy Clin Immunol Pract.* 2019;7(7):2252–2261. **8.** Fleming JT, Clark S, Camargo CA Jr., Rudders SA. *J Allergy Clin Immunol Pract.* 2015;3(1):57–62. **9.** Srisawat C, Nakponetong K, Benjasupattananun P, et al. *Asian Pac J Allergy Immunol.* 2016;34(1):38–43. **10.** McDonald R, Lorch U, Woodward J, et al. *Addiction.* 2018;113(3):484–493. **11.** Skulberg AK, Åsberg A, Khiabani HZ, et al. *Addiction.* 2019;114(5):859–867. **12.** Erdő F, Bors LA, Farkas D, et al. *Brain Res Bull.* 2018;143:155–170. **13.** Dworaczyk D and Hunt A. Presented at the American Academy of Allergy, Asthma & Immunology (AAAAI) National Conference, February 26–March 1, 2021, Virtual Annual Meeting

Study Events Flow Chart

Study Procedures	S	-1	Study in Each Period																				EOS or ET	FU
Days >	S	-1	1																					
Minutes >		C-I	P	-30	-20	-10	0	1	3	5	7	10	15	20	25	30	45	60	90	120	180	360		
Administrative Procedures	X	X																						
Safety Evaluations	X	X																						
Full Physical Examination	X	X																						
12-Lead Safety ECG	X		X													X		X			X	X	X	
Telemetry												X												
Vital Signs (HR and BP)*	X	X		X	X	X		X	X	X	X	X	X	X	X	X	X	X	X	X	X	X	X	
AE Monitoring																								X
Study Drug Administration / Pharmacokinetics • Epinephrine Administration • Blood for Epinephrine PK				X	X	X	X	X	X	X	X	X	X	X	X	X	X	X	X	X	X	X		
Intranasal Administration Tolerance Test	X																							
Intranasal Self-Administration Training**	X						X																	

Abbreviations: AE = Adverse event, BP = Blood pressure, C-I = Check-in, ECG = Electrocardiogram, EOS/ET = End of Study or early termination, FU = Follow-up, HR = Heart rate, P = Pre-dose, PK = Pharmacokinetics. *HR and BP measurements on Day 1 of each period were also used as PD markers. **At screening, subjects were trained in the correct use of the nasal spray for self-administration. In Treatment B, subjects were reminded of the correct technique prior to self-administration of the IN dose on Day 1.

Baseline Demographics

Trait	Category	Value
Sex n (%)	Male Female	29 (35%) 54 (65%)
Race	Asian Black or African American White White, American Indian/Alaska native White, Black or African American	4 (5%) 17 (20%) 55 (66%) 1 (1%) 6 (7%)
Ethnicity	Hispanic or Latino Not Hispanic or Latino	7 (8%) 76 (92%)
Age (yrs)	Mean (± SD) Median	36.4 ± 12.39 34.0
Weight (Kg)	Mean (± SD) Median	78.47 ± 13.22 77.9
Height (cm)	Mean (± SD) Median	173.0 ± 9.92 175.0
BMI	Mean (± SD)	26.15 ± 3.23

Median baseline-adjusted plasma epinephrine concentration-time profiles for IN self-administration compared to TP administration

	Self-administered* (Treatment B)		TP-administered* (Treatment A)				
Parameter	Geometric LSMs	n	Geometric LSMs	n	GMR (%)	90% Confidence Interval	Inter-subject CV%
AUC0-10,adj (pg*min/mL)	353.8	38	551.0	41	64.22	31.85-129.51	566.92
AUC0-20,adj (pg*min/mL)	1283	39	1519	41	84.51	56.92-125.48	144.46
AUC0-30,adj (pg*min/mL)	2302	39	2600	41	88.56	61.54-127.44	126.51
AUC0-60,adj (pg*min/mL)	5163	39	5570	41	92.69	67.24-127.78	105.02
AUC0-360,adj (pg*min/mL)	17970	31	16050	37	111.95	83.43-150.22	82.99
AUC0-t,adj (pg*min/mL)	14460	39	14440	41	100.17	75.14-133.55	90.33
AUC0-T_{max},adj (pg*min/mL)	1885	39	1716	41	109.84	72.95-165.41	153.27
AUC0-inf,adj (pg*min/mL)	21910	25	19120	32	114.58	83.95-156.38	79.01
Cmax,adj (pg/mL)	164.907	39	178.812	41	92.22	67.57-125.88	100.49
Cmax0-20,adj (pg/mL)	122.061	39	138.807	41	87.94	60.76-127.27	129.63

Treatment A: 6.6 mg epinephrine (0.11 mL x 60 mg/mL NDS1C) administered IN by a trained clinical personnel; **Treatment B:** 6.6 mg epinephrine (0.11 mL x 60 mg/mL NDS1C) self-administered IN; geometric least-squares means (LSMs) are calculated by exponentiating the LSMs from ANOVA. Geometric Mean Ratio (GMR) = 100*(test/reference); inter-subject CV% was calculated as 100 x square root(exp[MSE]-1), where MSE = Residual variance from ANOVA. Source: Table 14.2.3.20; program: /CA30843/sas_prg/pksas/adam_intext_statsmixed20.sas 08NOV2021 8:36; *First 6.6 mg IN spray self- versus TP-administered

2022
AAAAI ANNUAL MEETING

Conclusions

1. Comparable PK/PD/BA profiles for a single 6.6-mg IN dose of epinephrine were achieved with TP- or self-administration, suggesting that IN epinephrine can be effectively self-administered and may provide a useful alternative to IM injections.
2. In general, the PK parameters were similar for the 6.6-mg IN formulation whether TP- or self-administered, suggesting IN treatment can be considered equivalent regardless of the administrator.
3. No significant adverse changes in cardiovascular parameters were observed in patients treated with self-administered IN epinephrine compared to TP-administered IN epinephrine.
4. Self-administered IN epinephrine may provide a safe and effective alternative for the management of anaphylaxis, while allowing for convenient access to a second dose as required. Additional trials are ongoing.