Poster P005

Multicenter, Randomized Crossover Healthcare Professional Preference Study of Two-Dose Epinephrine Nasal Spray Versus Epinephrine Autoinjector

Jennifer Soosaar, PhD[1], Seda Japp, PhD[1], Sarah Johnstone, MA[1], Steven Rybicki, MSE[1], Erin Pohl, MS[1], Sarah Fairchild, PhD[1], Patricia Anderson, MSE[1], Adam Shames, MBA[1]

[1]Core Human Factors, Inc., Bala Cynwyd, PA, USA

Presented at the American College of Allergy, Asthma & Immunology National Conference, November 7–11, 2019, Houston, TX, USA

ABSTRACT

- **Introduction:** Preferences of healthcare professionals (HCPs) who treat patients with allergies were investigated between two devices that can be used in anaphylactic emergencies: the epinephrine autoinjector market leader (EAI) and a novel two-dose epinephrine nasal spray (ENS).
- **Methods:** Participants interacted with the devices in a simulated manner and then answered survey questions validated in advance of pivotal testing. The study involved 56 HCPs, including prescribers and clinical staff who train patients. The survey evaluated metrics including: portability, ease of learning, ease of use, overall preference, likelihood of recommending to others, and safety. Each question was analyzed with an exact binomial test (for statistical significance $p < .05$).
- **Results:** There was a statistically significant preference for ENS on all study metrics.
- **Conclusion:** Understanding preferences between devices that deliver comparable therapies can impact distribution of more ideal devices. Medical devices that are easier to use are typically safer and more effective at delivering the correct dose of medication. This study indicates that the ENS device is perceived to be easier to use in several ways and may be more likely to be carried by patients due to its size and portability. When considering that the most common cause of death from food allergies is delayed epinephrine administration,[1] prescription and use of ENS over the specific EAI used in this study might provide significant advantages and decrease the incidence of anaphylactic deaths. The HCP data presented herein align with patient preferences previously identified by the authors.

INTRODUCTION

- Immediate administration of epinephrine, currently most commonly self-administered in patients via intramuscular or subcutaneous injection with an autoinjector, is the first-line therapy in the treatment of a severe allergic reaction (anaphylaxis)[2]
- Delayed treatment with epinephrine in anaphylaxis can lead to complications and increased rates of hospitalizations and, potentially, death[1–3]
- Both patients and healthcare professionals (HCPs) show inadequate understanding of correct autoinjector use[4]
- Proper training of HCPs on how to communicate and overcome barriers related to epinephrine use and a clear understanding of patient preferences related to device options allow for more effective communication about treatment protocols, which may in turn improve patient adherence and treatment during anaphylaxis[5,6]

AIM

- To investigate HCP preferences between two medical devices that could be used to deliver epinephrine in anaphylactic emergencies: the novel two-dose epinephrine nasal spray (ENS) and the epinephrine autoinjector (EAI) market leader

METHODS

- This was a multicenter, randomized, crossover preference study of ENS versus EAI

Study devices (Figure 1)

- The devices used in the study were:
 - Empty but fully functional ENS devices. Each ENS device contains two equivalent doses; therefore, users only have to carry one device
 - EAI trainer devices, which do not contain the drug or a needle. The EAI contains one dose; therefore, users have to carry two devices

Figure 1. ENS device (left) and EAI (right) used in the study[a]

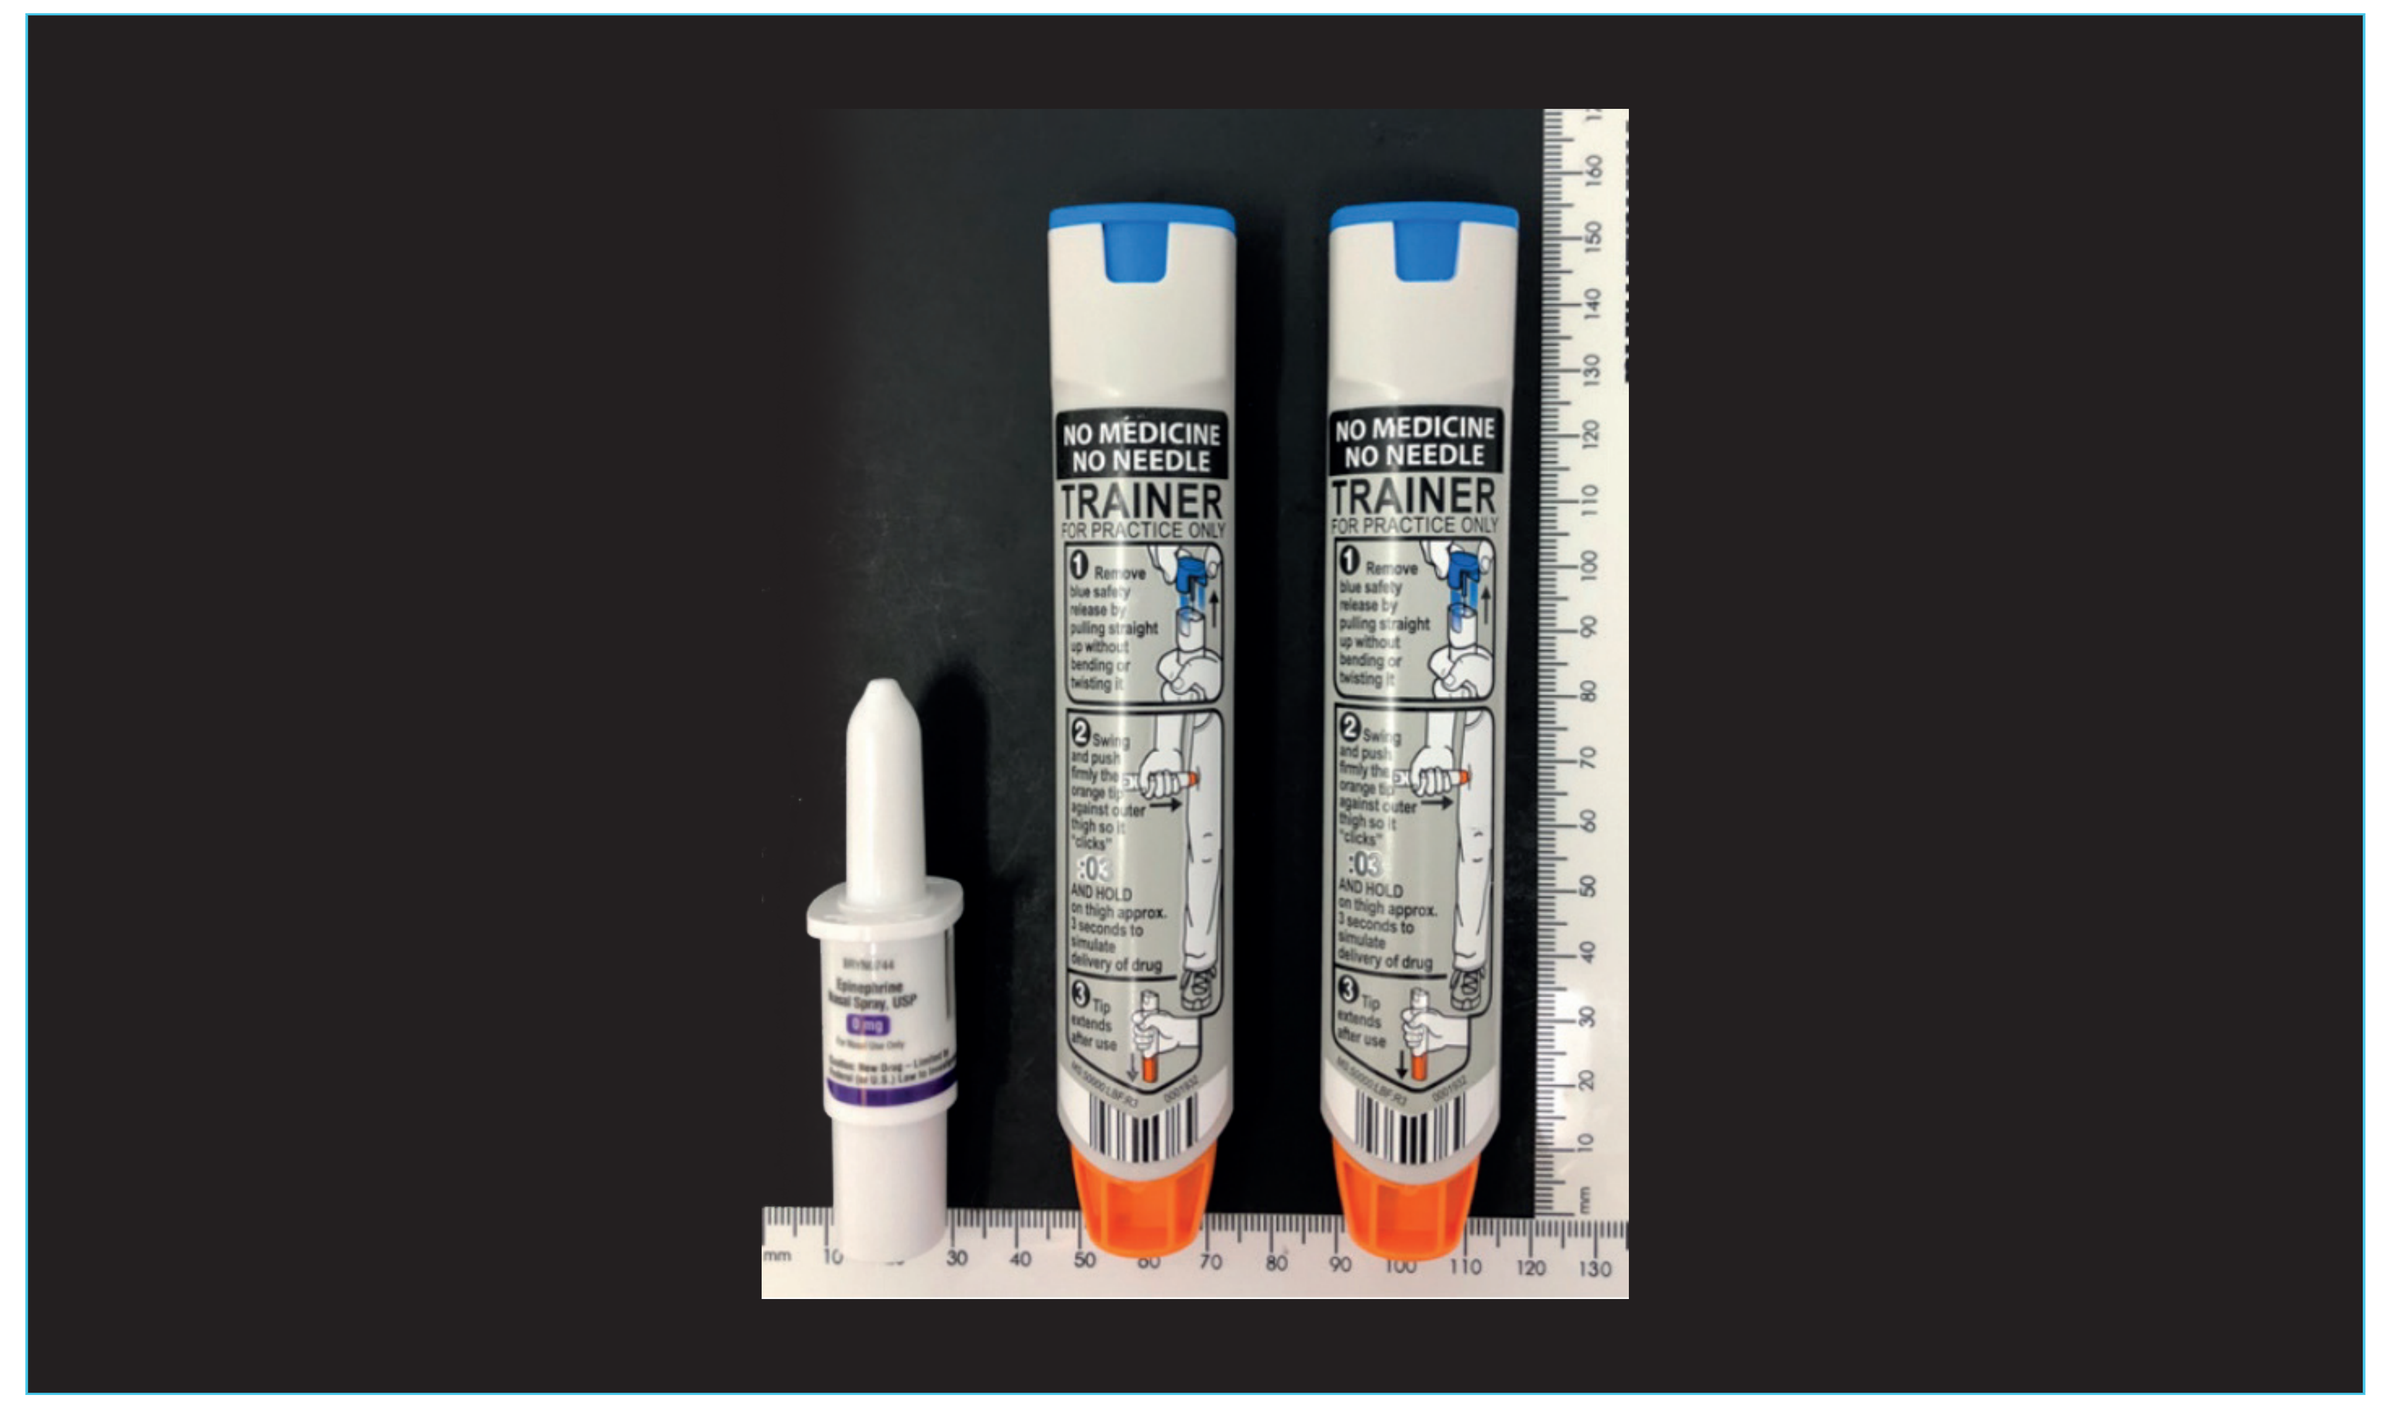
NO MEDICINE
NO NEEDLE
TRAINER
FOR PRACTICE ONLY
NO MEDICINE
NO NEEDLE
TRAINER
FOR PRACTICE ONLY

[a]Image depicts equivalent doses (one ENS device equals two EAI devices)
EAI, epinephrine autoinjector; ENS, epinephrine nasal spray

- HCPs simulated use of the ENS by holding the device in the air and pressing the plunger, as this is how HCPs are expected to demonstrate use of the nasal spray device to patients
- HCPs simulated use of the EAI by removing the blue safety cap, pushing the orange tip that houses the needle into their thigh until it clicked, holding the device in that position for 3 seconds, and then releasing the pressure to simulate removing the needle

Questionnaire development

- To assess HCP preference, an eight-item, forced-choice questionnaire was developed based on differences between the ENS device and EAI for epinephrine administration
 - Preference was assessed on various metrics, including ease of patient training, ease of use, portability, safety, comfort, size, and emergency use
- Three answer choices were provided for each survey question: ENS, EAI, or no preference
- This questionnaire was iteratively refined in a pilot study of 13 participants (7 patients and 6 HCPs) and finalized in advance of pivotal preference testing

Pivotal preference study

- Two user groups participated in this preference study:
 - Patients aged ≥11 years diagnosed with severe allergies and prescribed an EAI
 - Results from the patient participant were presented recently[7]
 - HCPs who treat patients with severe allergies; this group included prescribers of epinephrine and clinical staff who train patients on use of epinephrine
- HCPs from New York, Massachusetts, Georgia, Texas, and California participated in the study
- All participants provided written consent before their session and were compensated for participating
- Participants attended a one-on-one session with a Moderator during which they were trained on how to use both devices
 - Participants observed a demonstration of the first device and then simulated use of the first device on their own. They then observed a demonstration of the second device and then simulated use of the second device on their own. Presentation of the devices was counterbalanced between participants
 - After using both devices, participants completed the eight-item questionnaire
- This study was reviewed and approved by the Core Human Factors, Inc., Independent Review Board, which is registered with the US Department of Health and Human Services

Statistical analysis

- Each question was analyzed separately for HCPs with an exact binomial test using the binomial test function in the statistical software “R” (The R Foundation, Vienna, Austria)
- A probability value (p-value) of $p = .05$ was used to evaluate statistical significance

RESULTS

- Fifty-six HCPs participated in the study (**Table 1**)

Table 1. Number of HCPs by participant type

User Group	n
Primary care physician	15
Allergy/immunology physician	15
Primary care nurse/trainer	16
Allergy/immunology nurse/trainer	10
Total	**56**

HCPs, healthcare professionals

- For all metrics evaluated, the survey results indicated a statistically significant preference for the ENS device over the EAI (**Table 2; Figures 2** and **3**)

Table 2. HCP preference for ENS device

Question	Preference for ENS Device, n (%) (N=56)	*p*-value
Which device do you think a patient would be more likely to carry with them in daily life?	50 (89.3)	<.001
Which device do you think would be easier for patients to learn how to use?	43 (76.8)	<.001
Which device is easier to use?	46 (82.1)	<.001
Which device do you prefer overall?	38 (67.9)	<.001
Which device would you recommend to patients?	37 (66.1)	<.001
Which device do you think would be safer for a patient to use?	39 (69.6)	<.001
Which device do you prefer based on the size of the device?	46 (82.1)	<.001
Which device do you think a patient would be more likely to use in a real emergency?	29 (51.8)	.003

EAI, epinephrine autoinjector; ENS, epinephrine nasal spray

- Regarding device portability, 89.3% of HCPs expected that patients would prefer to carry the ENS device compared to the EAI in daily life ($p < .001$)
- Regarding device teachability, 76.8% of HCPs indicated that it would be easier for patients to learn how to use the ENS device than the EAI ($p < .001$)
- Regarding device safety, 69.6% of HCPs indicated that the ENS device seemed safer for a patient to use than the EAI ($p < .001$)
- Regarding device use in emergency, 51.8% of HCPs expected that patients would prefer to use the ENS device in a real emergency instead of the EAI ($p < .003$)
- Regarding device recommendation, 66.1% of HCPs indicated they would recommend the ENS device to patients over the EAI ($p < .001$)
- Regarding device size, 82.1% of HCPs preferred the ENS size overall compared to the EAI size ($p < .001$)
- Regarding ease of use, 82.1% if HCPs preferred the ENS ease of use over the EAI ($p < .001$)
- Regarding overall preference, 67.9% of HCPs preferred the ENS device overall over the EAI ($p < .001$)

Figure 2. Device preference of HCPs

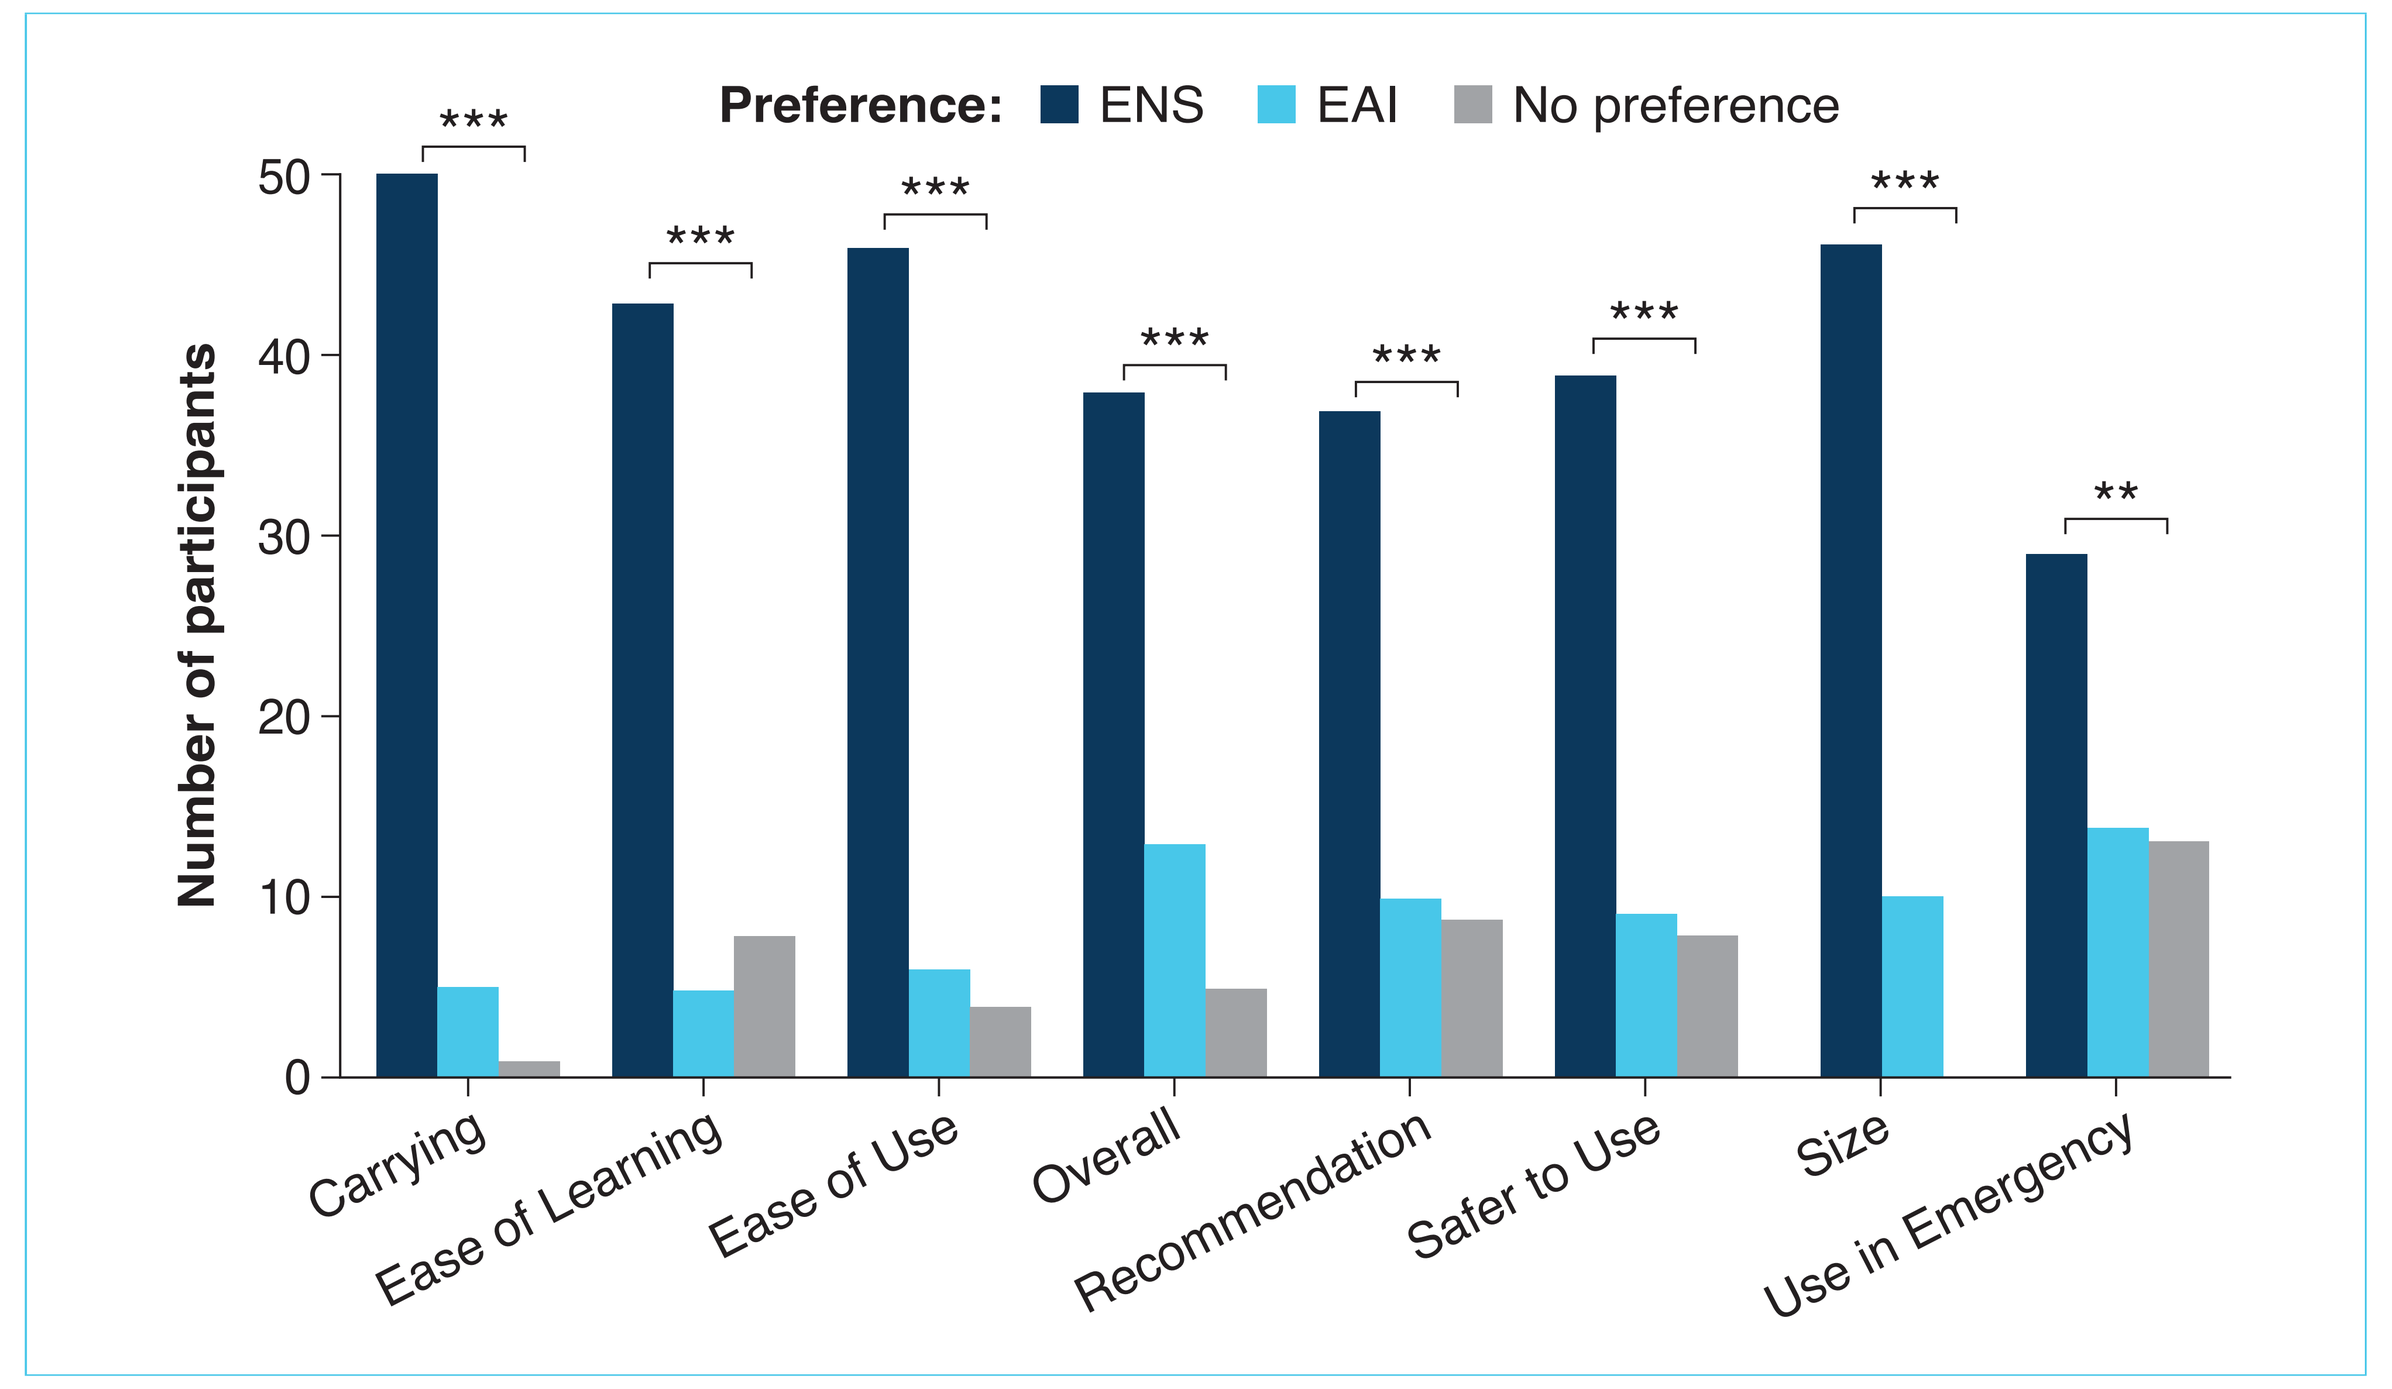
Preference: ENS EAI No preference
Number of participants
0
10
20
30
40
50

**
Carrying
Ease of Learning
Ease of Use
Overall
Recommendation
Safer to Use
Size
Use in Emergency

$p < .01$, *$p < .001$
EAI, epinephrine autoinjector; ENS, epinephrine nasal spray; HCPs, healthcare professionals

Figure 3. Preference of HCPs for the ENS over the EAI

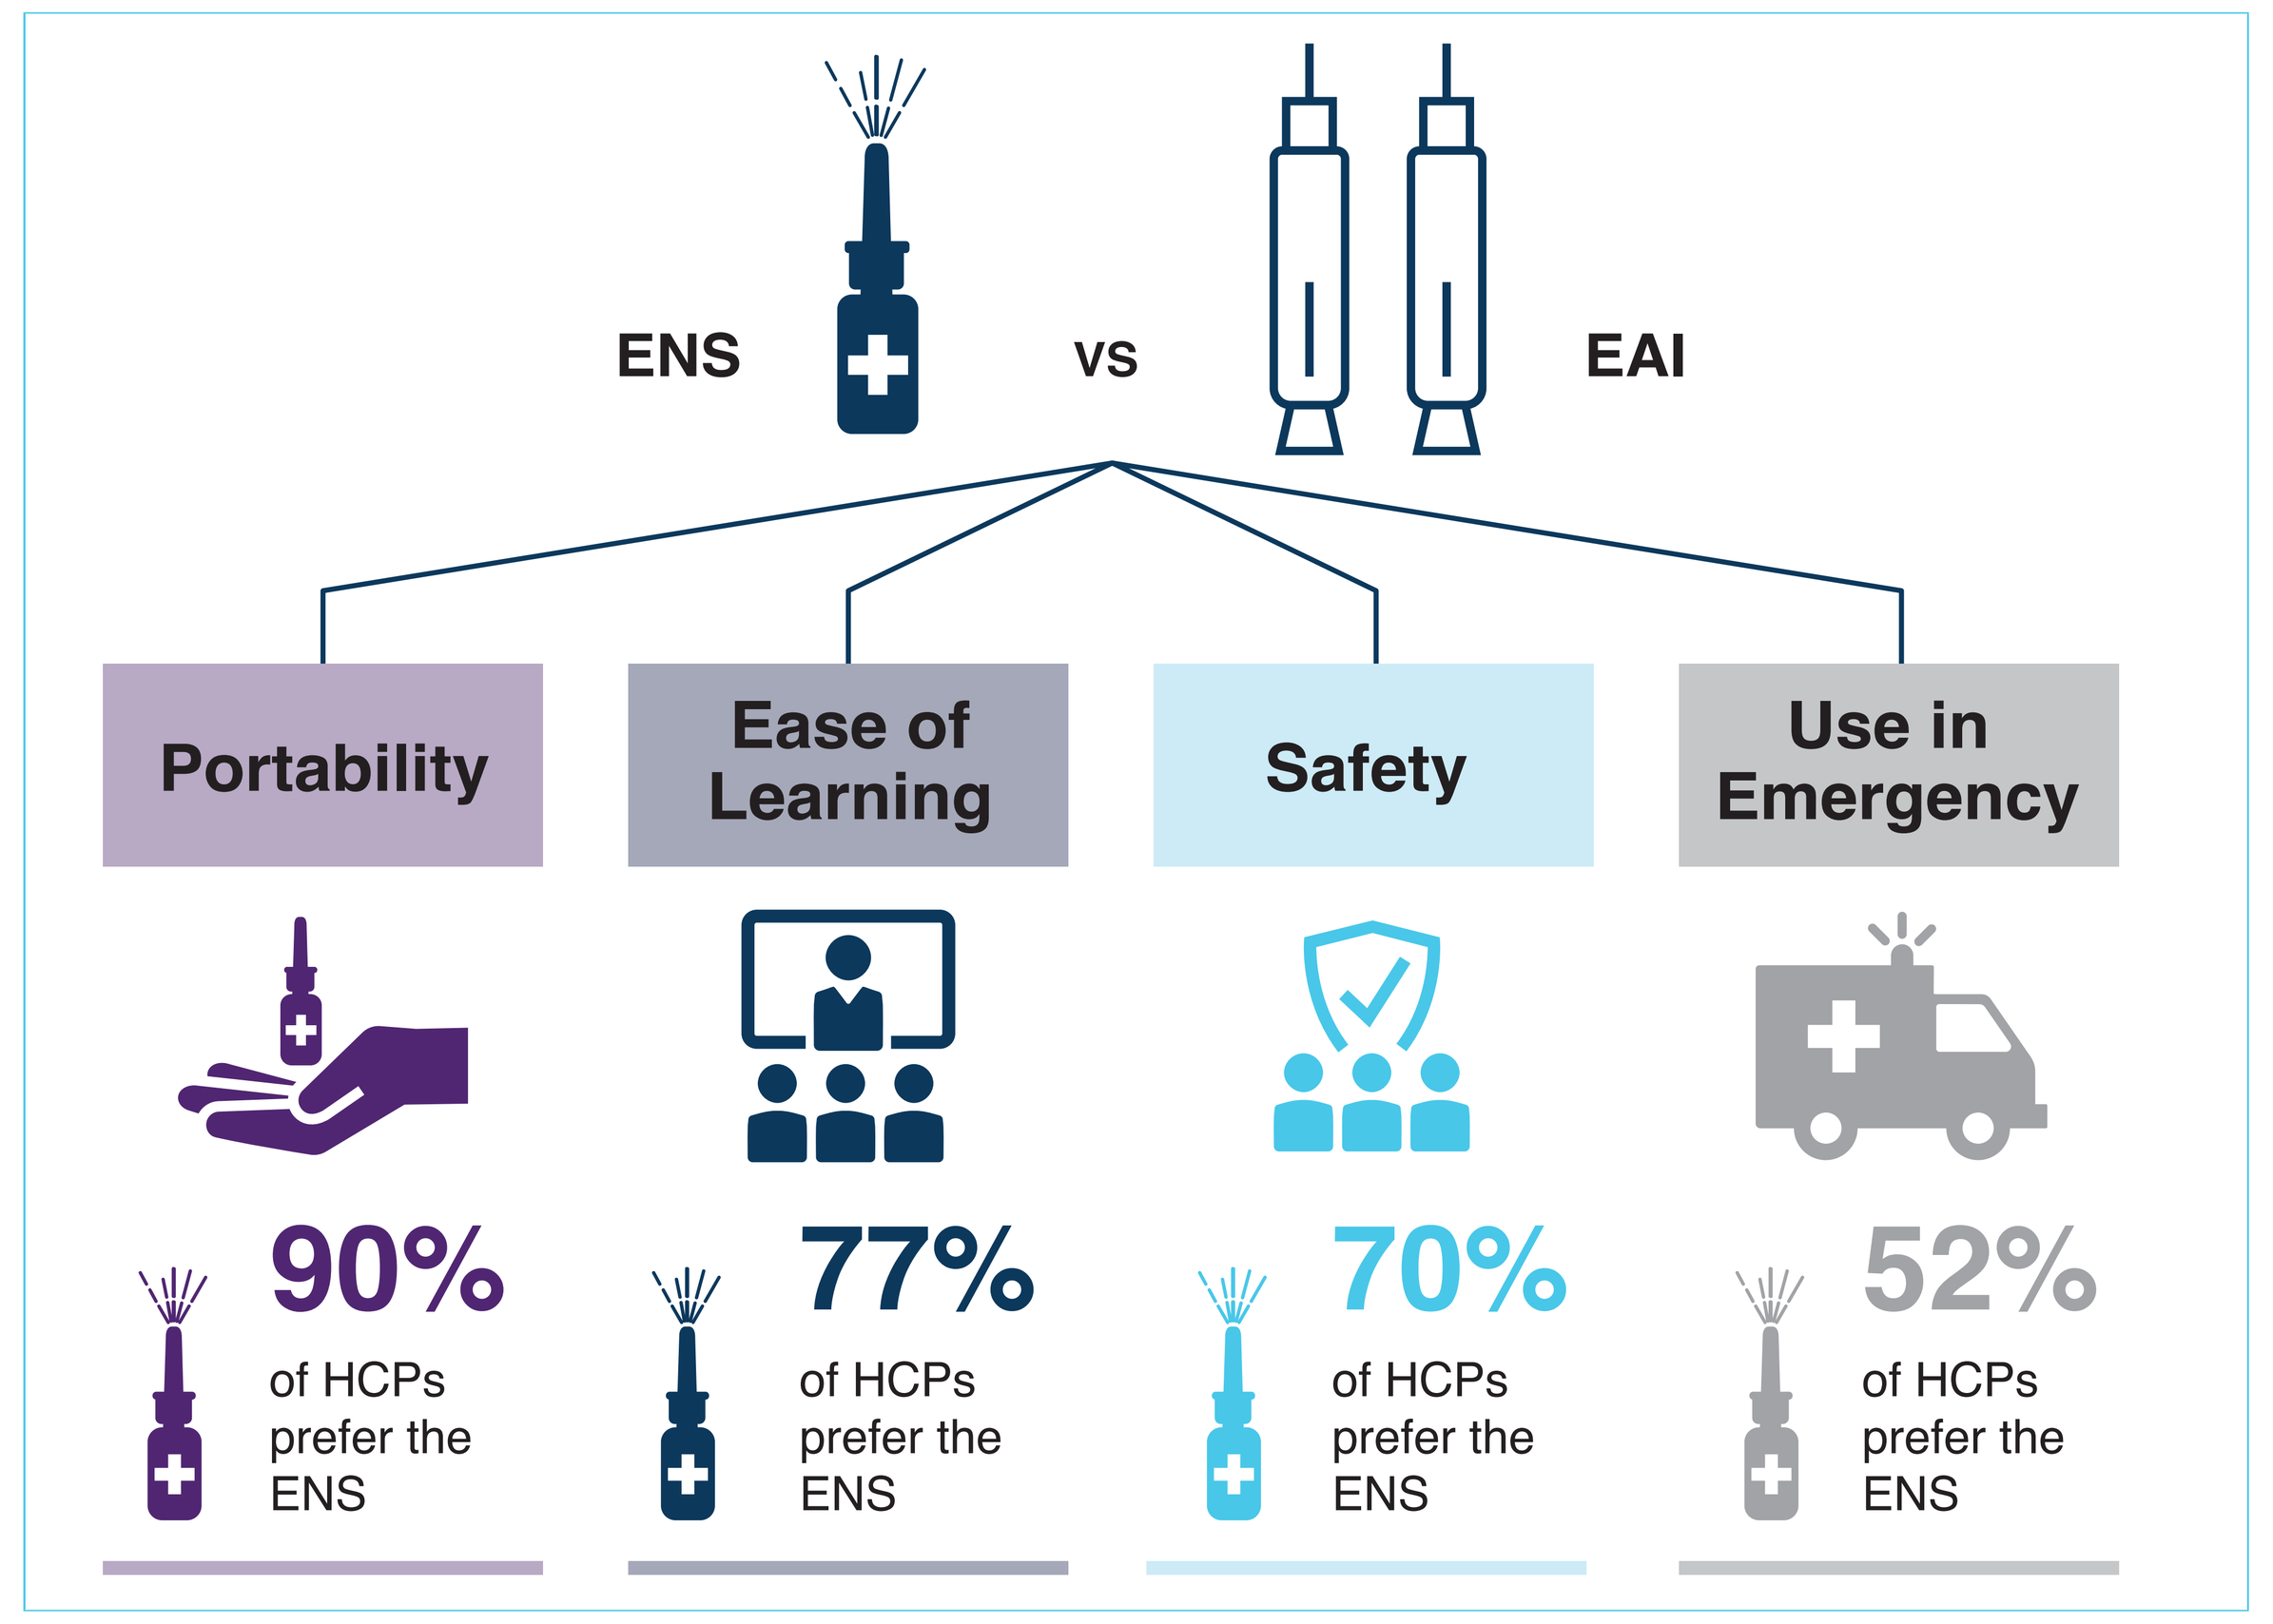
ENS
VS
EAI
Portability
Ease of Learning
Safety
Use in Emergency
90%
of HCPs prefer the ENS
77%
of HCPs prefer the ENS
70%
of HCPs prefer the ENS
52%
of HCPs prefer the ENS

HCPs, healthcare professionals

CONCLUSIONS

1. HCP preferences and their expectations of patient preferences can influence prescribing behavior, which in turn affects patient outcomes
2. HCPs preferred the ENS device over the EAI for all metrics evaluated, including portability, ease of teaching, ease of use, overall preference, likelihood of recommending to patients, safety, size, and likelihood of a patient using in a real emergency
3. Use of the ENS device instead of the EAI for the treatment of anaphylaxis may increase patient adherence and potentially reduce hospitalizations and anaphylactic deaths

References

1. Chooniedass R, et al. *Ann Allergy Asthma Immunol.* 2017;119(2):108-110.
2. Posner LS, Camargo CA Jr. *Drug Healthc Patient Saf.* 2017;9:9-18.
3. Bellanti JA, Settipane RA. *Allergy Asthma Proc.* 2014;35(3):195-196.
4. Ewan P, et al. *Clin Exp Allergy.* 2016;46(12):1621-1622.
5. Mahoney B, et al. *Immun Inflamm Dis.* 2019;7(3):214-228.
6. Portnoy J, et al. *J Allergy Clin Immunol Pract.* 2019;7(7):2252-2261.
7. Soosaar J, et al. Presented at: American Academy of Pediatrics National Conference 2019; New Orleans, LA.

ACKNOWLEDGMENTS AND DISCLOSURES

This research was supported by Bryn Pharma, LLC. Editing assistance was provided by Caryne Craige, PhD, of Fishawack Communications Inc.; funding for this assistance was provided by Bryn Pharma, LLC.